Poster #4083

In vivo efficacy of LGR5-targeting CAR-T cell therapies developed for the treatment of colorectal cancer

Dylan J. McPeake[1], Timona S. Tyllis[1], Caitlin A. Abbott[1], Jade Foeng[1], Veronika Bandara[3], Batjargal Gundsambuu[3], Elaheh Rohani-Rad[1], Silvana Napoli[2], Timothy Sadlon[2], Iain Comerford[1], Simon C. Barry[2,3] and Shaun R. McColl[1, 4].

[1] Chemokine Biology Laboratory, Department of Molecular and Biomedical Science, The University of Adelaide, Adelaide, South Australia, 5005.
[2] Department of Paediatrics, Women's and Children's Health Network, North Adelaide, South Australia, 5006.
[3] Molecular Immunology, Robinson Research Institute, The University of Adelaide, North Adelaide, South Australia, 5006.
[4] Carina Biotech, Innovation & Collaboration Centre, University of South Australia, Adelaide, South Australia, 5000.

Introduction

While chimeric antigen receptor (CAR)-T cells have demonstrated significant efficacy in the treatment of haematological malignancies, there are many challenges to overcome in developing CAR-T cell therapies against solid tumours. In particular, identification of target antigens in solid tumours remains a significant hurdle. Leucine-rich repeat-containing G-protein coupled receptor (LGR5) is a cancer stem cell marker, which mediates important roles in tumor initiation and metastasis (Carmon et al., 2011).

Previously, we have demonstrated that LGR5 is expressed by numerous human cancers, including colorectal, ovarian, hepatic, brain, and pancreatic cancer (AACR 2022). Additionally, LGR5-targeting CAR-T cells effectively kill LGR5-expressing cancer cells *in vitro* (AACR 2023 poster #4085). Upregulated LGR5 expression in diverse malignant cell contexts (McClanahan et al., 2006) positions this surface receptor as a promising target for CAR-T cell therapy.

The aim of this study was to determine the efficacy of LGR5-targeting CAR-T cells against colorectal cancer *in vivo*.

Summary

The *in vivo* efficacy of LGR5-targeting CAR-T cells was evaluated in two human colorectal cancer xenograft models, LoVo and LIM1215. LGR5-targeting CAR-T cells displayed significant anti-tumor activity against established tumors and upon rechallenge. Analysis of *in vivo* spatiotemporal distribution demonstrated that LGR5-targeting CAR-T cells do not persist in off-target organs by d28 post-tumor inoculation and are well-tolerated under the conditions tested. In contrast, these cells accumulate at the tumor site until tumor elimination.

Together, these results provide a foundation for furthering the development of LGR5-targeting CAR-T cells for use in the clinic against colorectal cancers and other solid tumors.

Acknowledgements

This research was funded by Carina Biotech. D.J.M. is the recipient of a scholarship co-funded by Carina Biotech and The University of Adelaide.

References:

Carmon, K. S., Gong, X., Lin, Q., Thomas, A., & Liu, Q. (2011). R-spondins function as ligands of the orphan receptors LGR4 and LGR5 to regulate Wnt/β-catenin signaling. *Proceedings of the National Academy of Sciences*, *108*(28), 11452–11457.

McClanahan, T., Koseoglu, S., Smith, K., Grein, J., Gustafson, E., Black, S., Kirschmeier, P., & Samatar, A. A. (2006). Identification of overexpression of orphan G protein-coupled receptor GPR49 in human colon and ovarian primary tumors. *Cancer Biology & Therapy*, *5*(4), 419–426.

Results

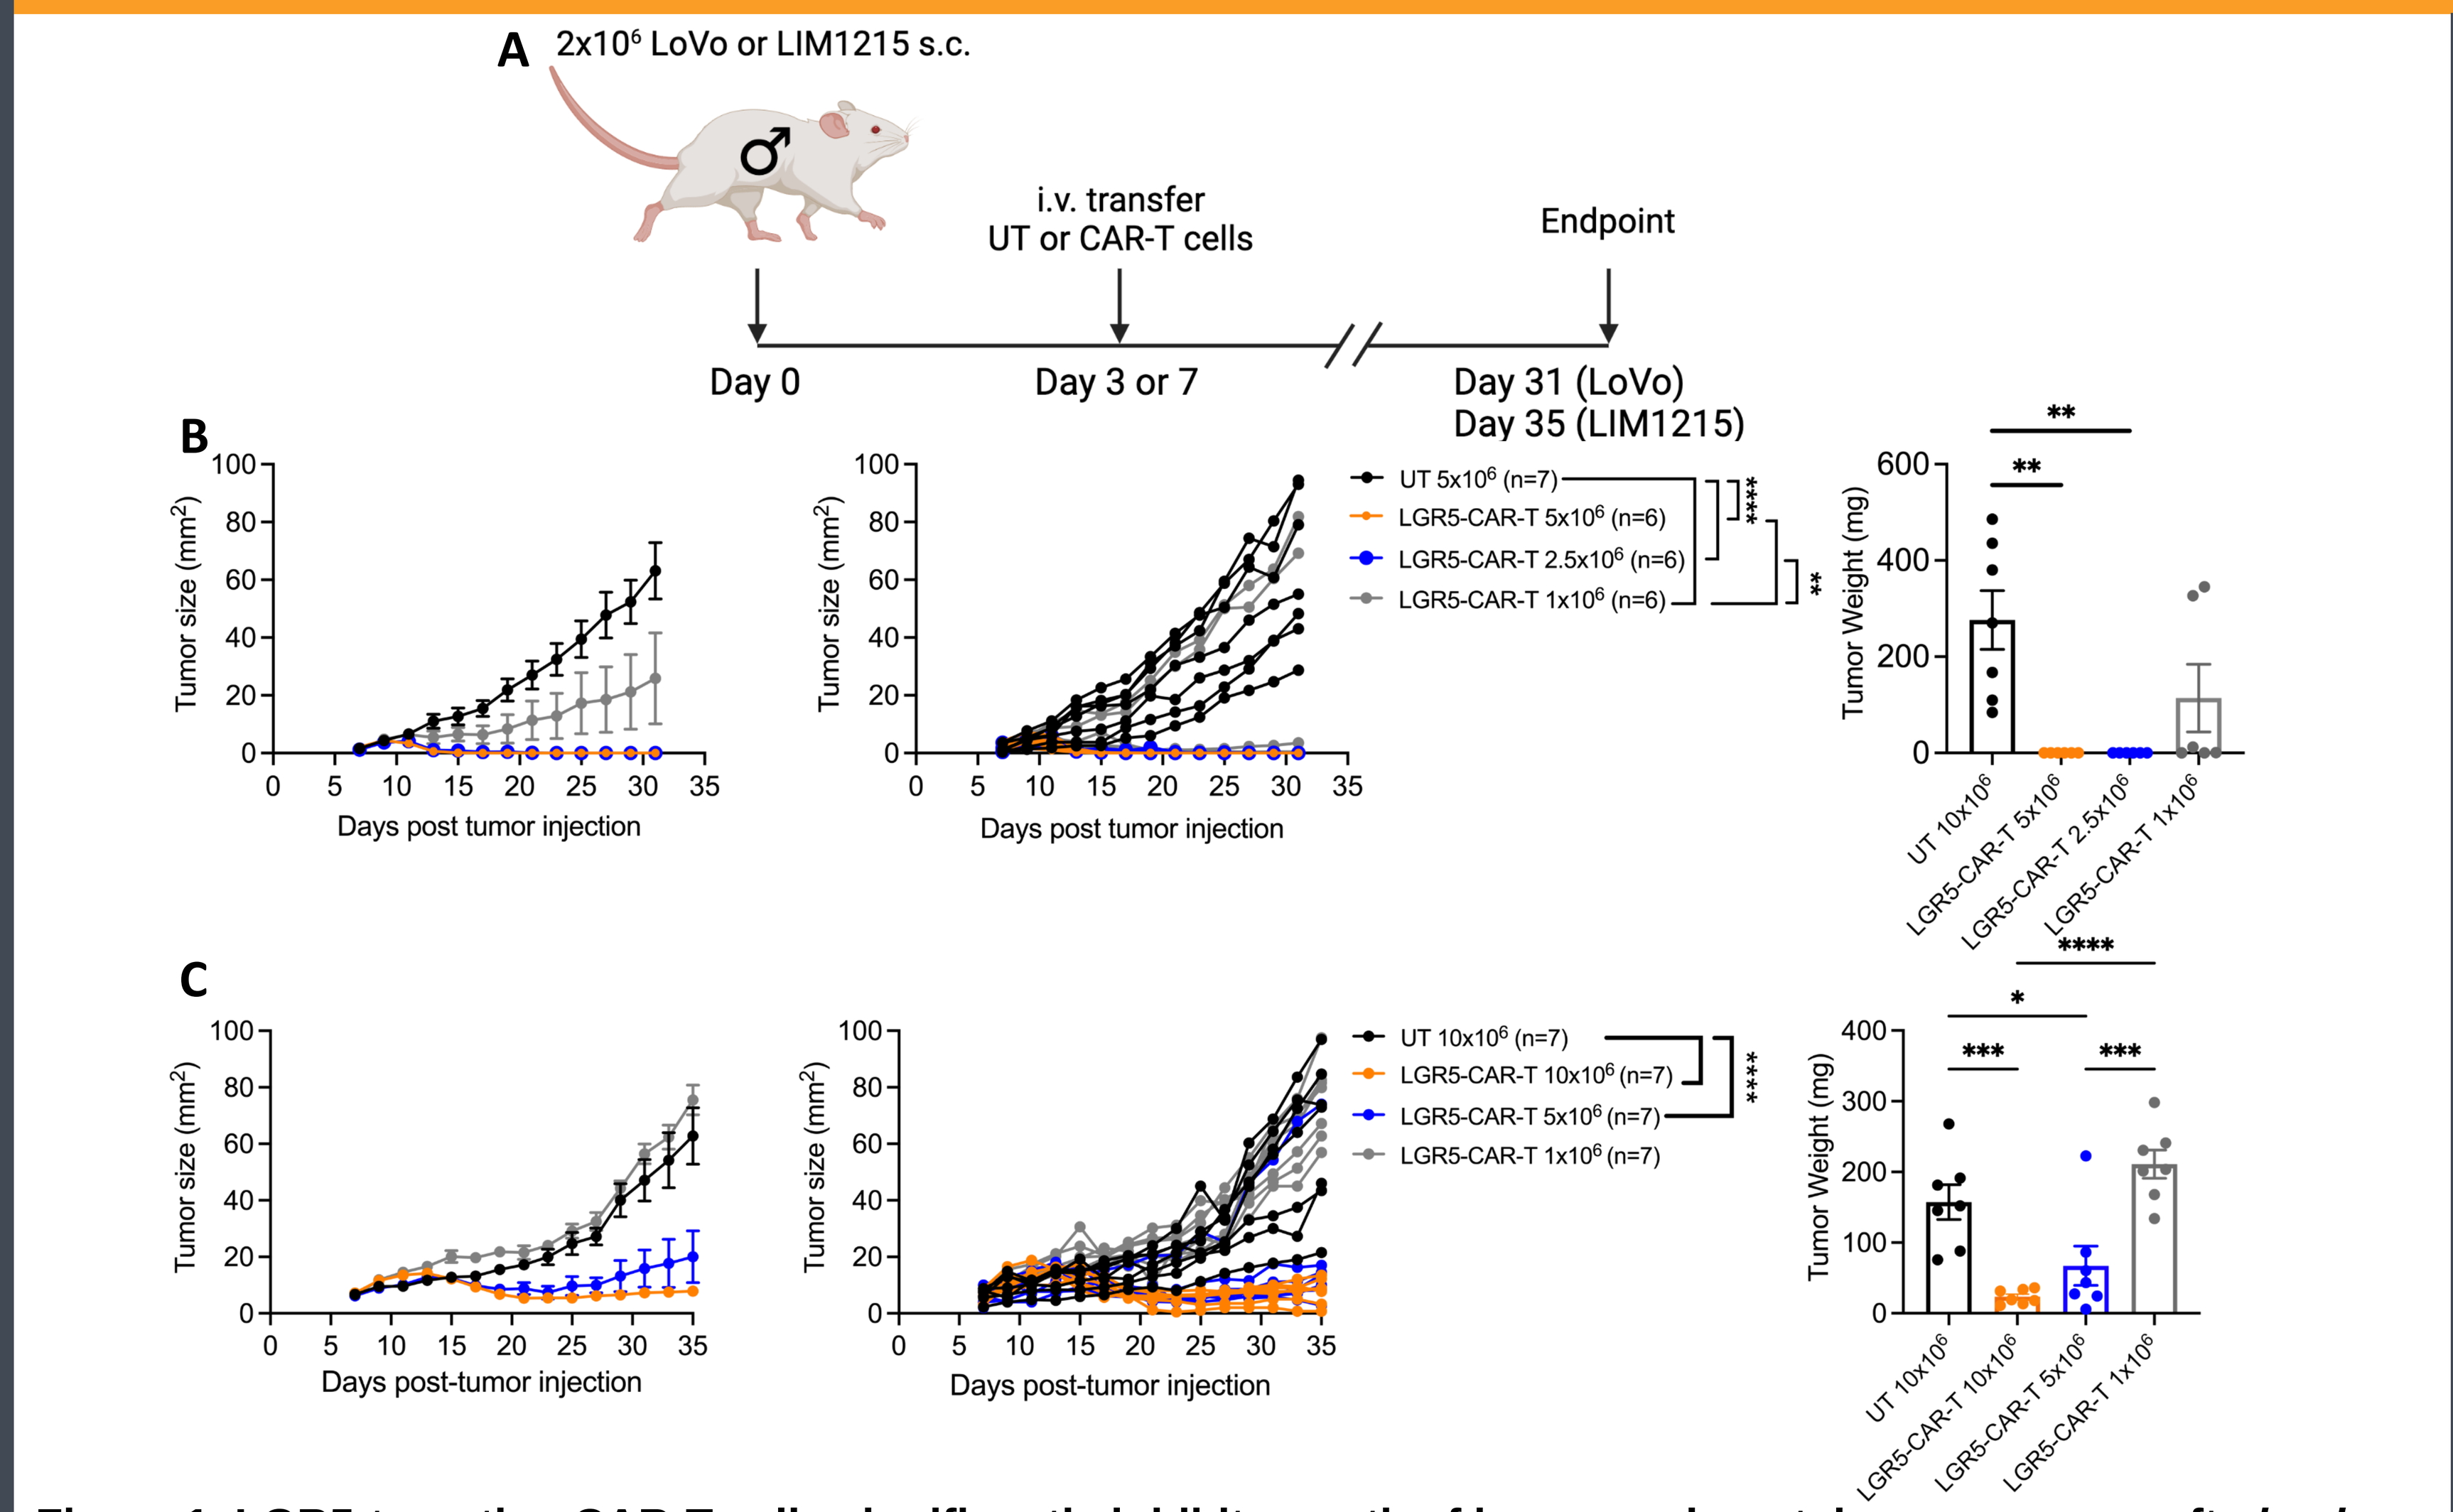

A
2x10⁶ LoVo or LIM1215 s.c.
i.v. transfer UT or CAR-T cells
Endpoint
Day 0
Day 3 or 7
Day 31 (LoVo)
Day 35 (LIM1215)
B
C
Tumor size (mm²)
Days post tumor injection
Days post-tumor injection
UT 5x10⁶ (n=7)
LGR5-CAR-T 5x10⁶ (n=6)
LGR5-CAR-T 2.5x10⁶ (n=6)
LGR5-CAR-T 1x10⁶ (n=6)
UT 10x10⁶ (n=7)
LGR5-CAR-T 10x10⁶ (n=7)
LGR5-CAR-T 5x10⁶ (n=7)
LGR5-CAR-T 1x10⁶ (n=7)
Tumor Weight (mg)

Figure 1. LGR5-targeting CAR-T cells significantly inhibit growth of human colorectal cancer xenografts *in vivo*. LGR5-targeting CAR-T cell efficacy was tested against human colorectal xenograft models, LoVo and LIM1215. $2x10^6$ tumor cells were injected subcutaneously (s.c.) into male NOD-*scid* IL2Rγ^{null} (NSG) mice, and at 3-7-days post-tumor injection, different doses of UT (untransduced) control and CAR-T cells were transferred intravenously (i.v.). LGR5-targeting CAR-T cells harbouring either a medium or short length hinge were tested in the LoVo or LIM1215 model, respectively. **(A)** Experiment schematic. **(B)** Grouped (left) and individual (right) LoVo tumor growth curves. **(C)** Endpoint LoVo tumor weights. **(D)** Grouped (left) and individual (right) LIM1215 tumor growth curves. **(E)** Endpoint LIM1215 tumor weights. Data in **(B,D)** two-way ANOVA with Bonferroni's post-test , **(C,E)** one-way ANOVA; Bonferroni's post-test. All data presented as mean ± SEM; ** p≤0.01, ***p≤0.001, ****p≤0.0001.

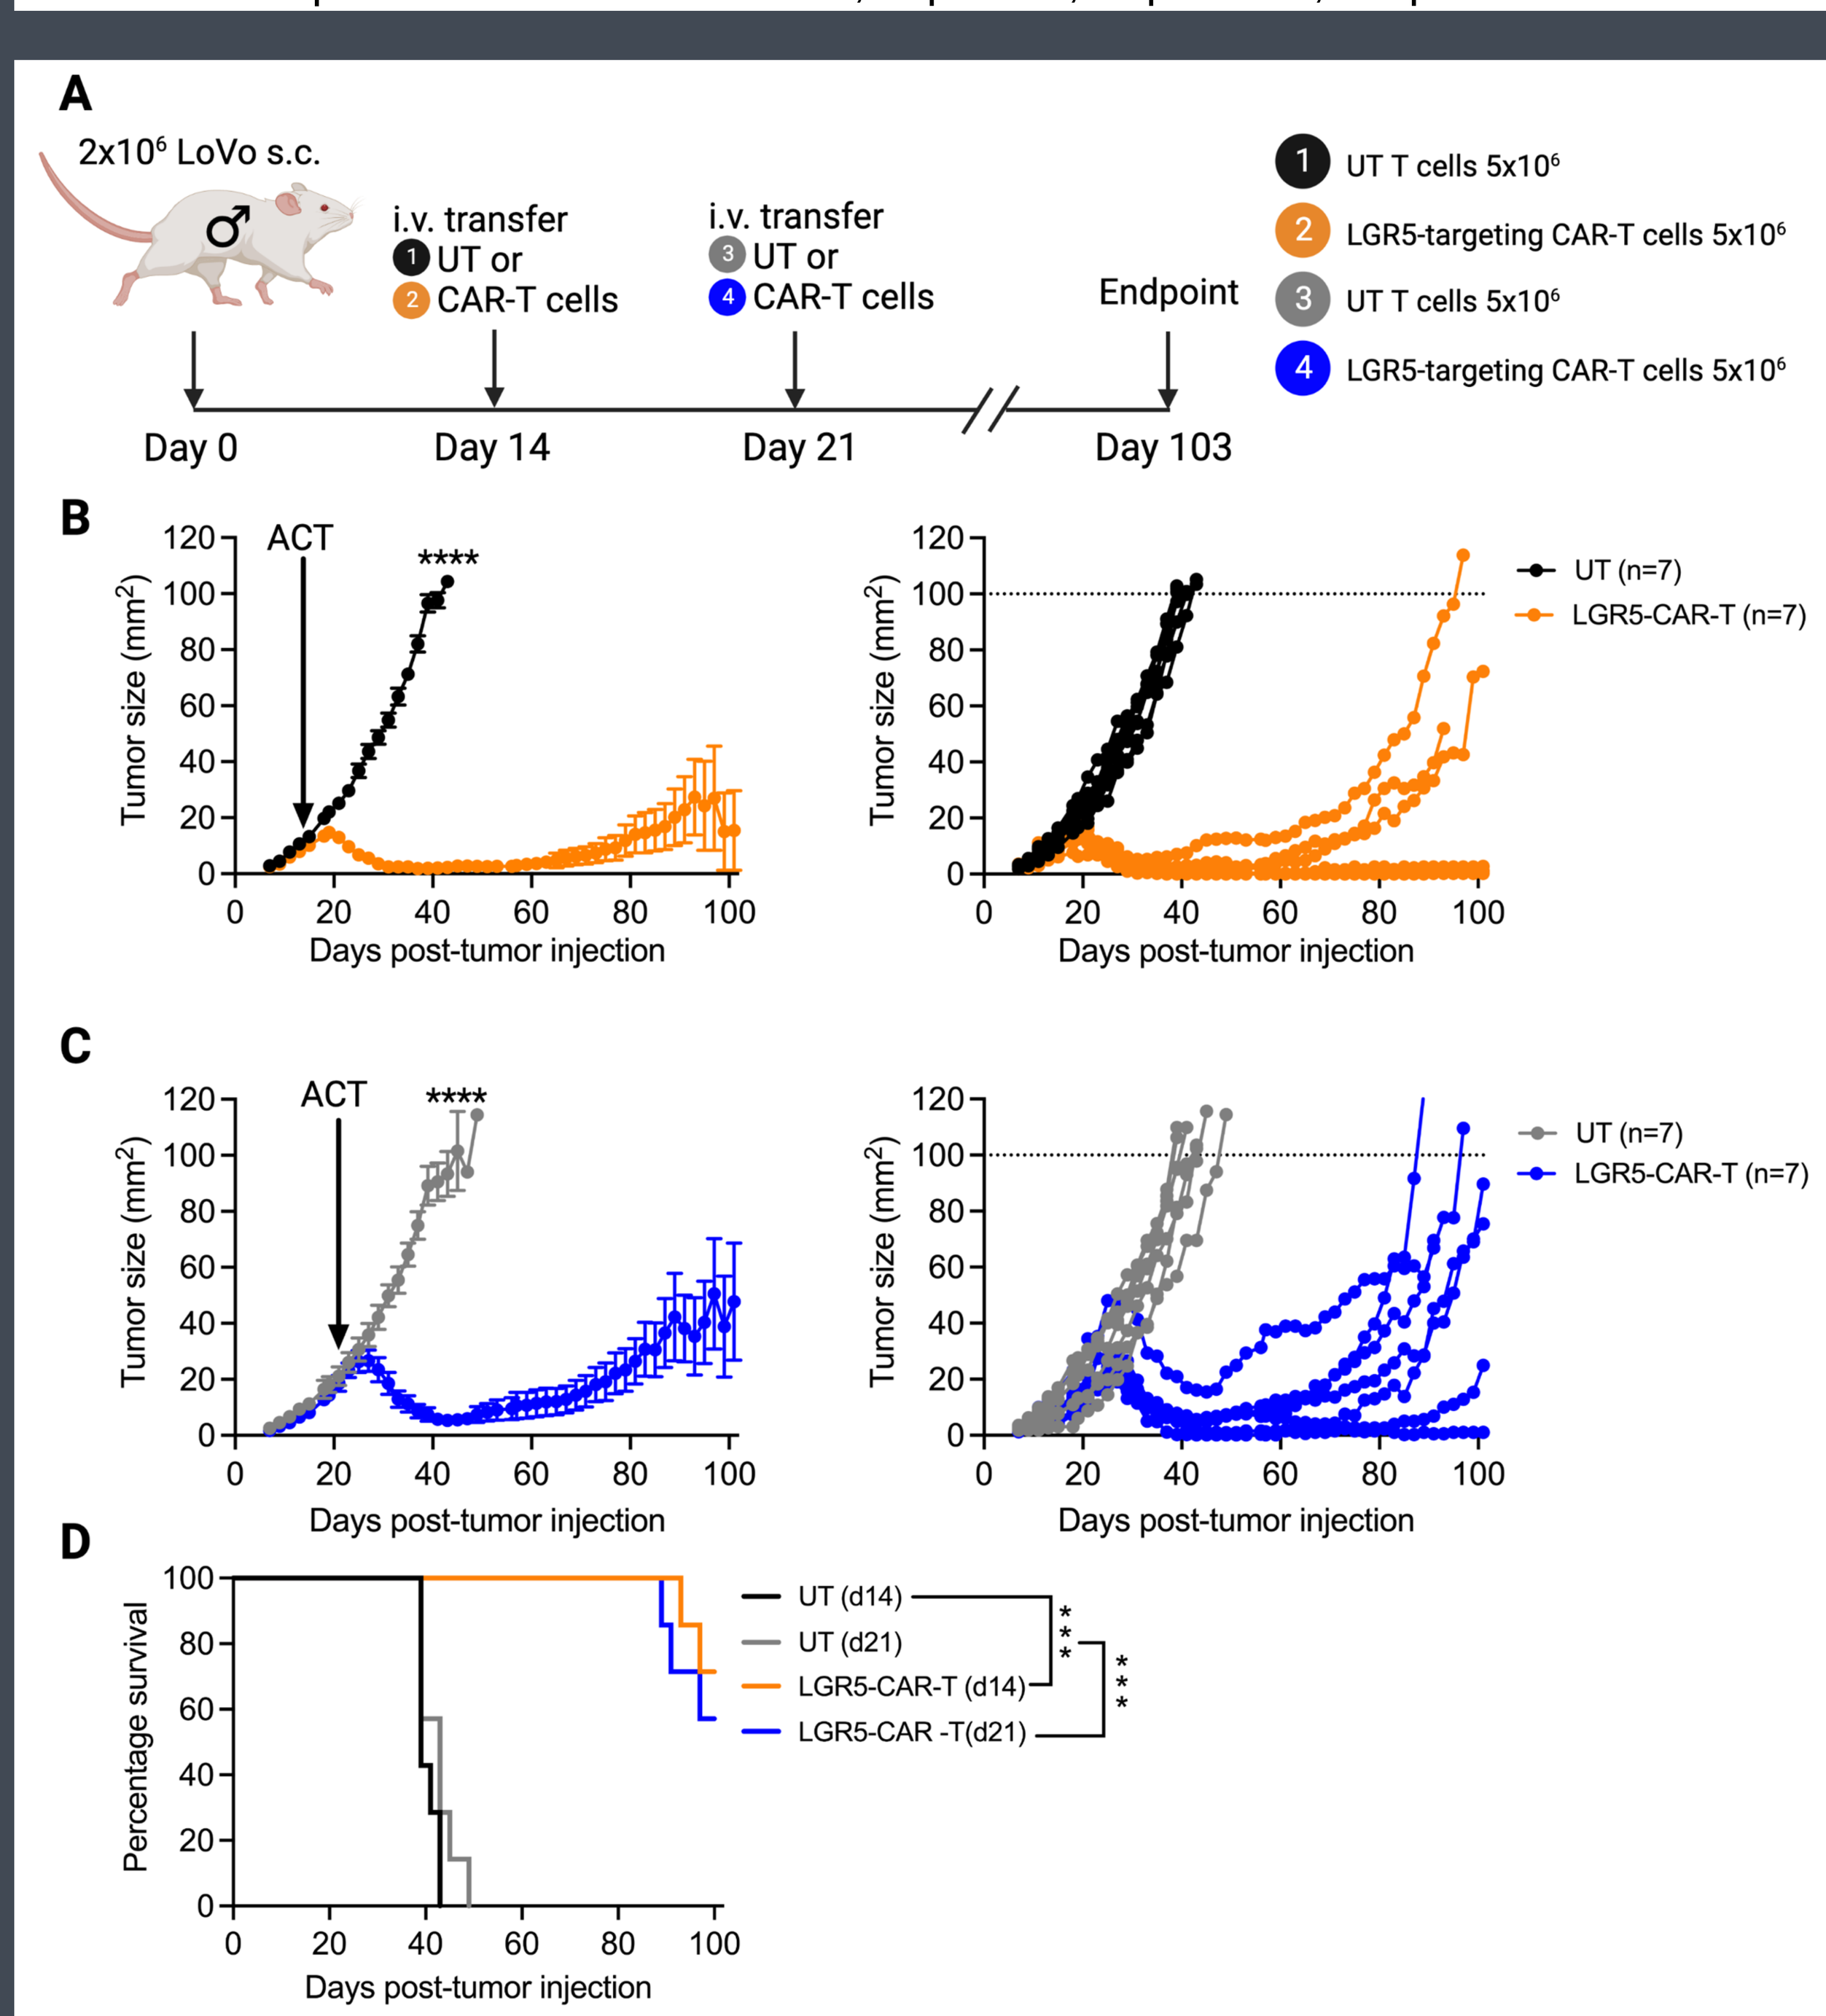

A
2x10⁶ LoVo s.c.
i.v. transfer 1 UT or 2 CAR-T cells
i.v. transfer 3 UT or 4 CAR-T cells
Endpoint
Day 0
Day 14
Day 21
Day 103
1 UT T cells 5x10⁶
2 LGR5-targeting CAR-T cells 5x10⁶
3 UT T cells 5x10⁶
4 LGR5-targeting CAR-T cells 5x10⁶
B
C
D
ACT
Tumor size (mm²)
Days post-tumor injection
UT (n=7)
LGR5-CAR-T (n=7)
Percentage survival
UT (d14)
UT (d21)
LGR5-CAR-T (d14)
LGR5-CAR -T(d21)

Figure 2. LGR5-targeting CAR-T cells supress the growth of established LoVo tumors and prolong survival. $5x10^6$ UT control or LGR5-targeting CAR-T cells were transferred i.v. on day 14 and 21 post-tumor injection in LoVo-bearing male NSG mice. **(A)** Experiment schematic. Grouped (left) and individual (right) LoVo tumor growth curves in mice that received adoptive cell transfer (ACT) on **(B)** day 14 or **(C)** day 21 post-tumor injection as indicated by the black arrow. Dotted lines represent the $100mm^2$ tumor area euthanasia threshold. **(D)** Kaplan-Meier survival curve. Data in **(B,C)** represented as the mean ± SEM; two-way ANOVA with Bonferroni's post-test and data in (C) represented as grouped data; Mantel-Cox test; ***p≤0.001, ****p≤0.0001.

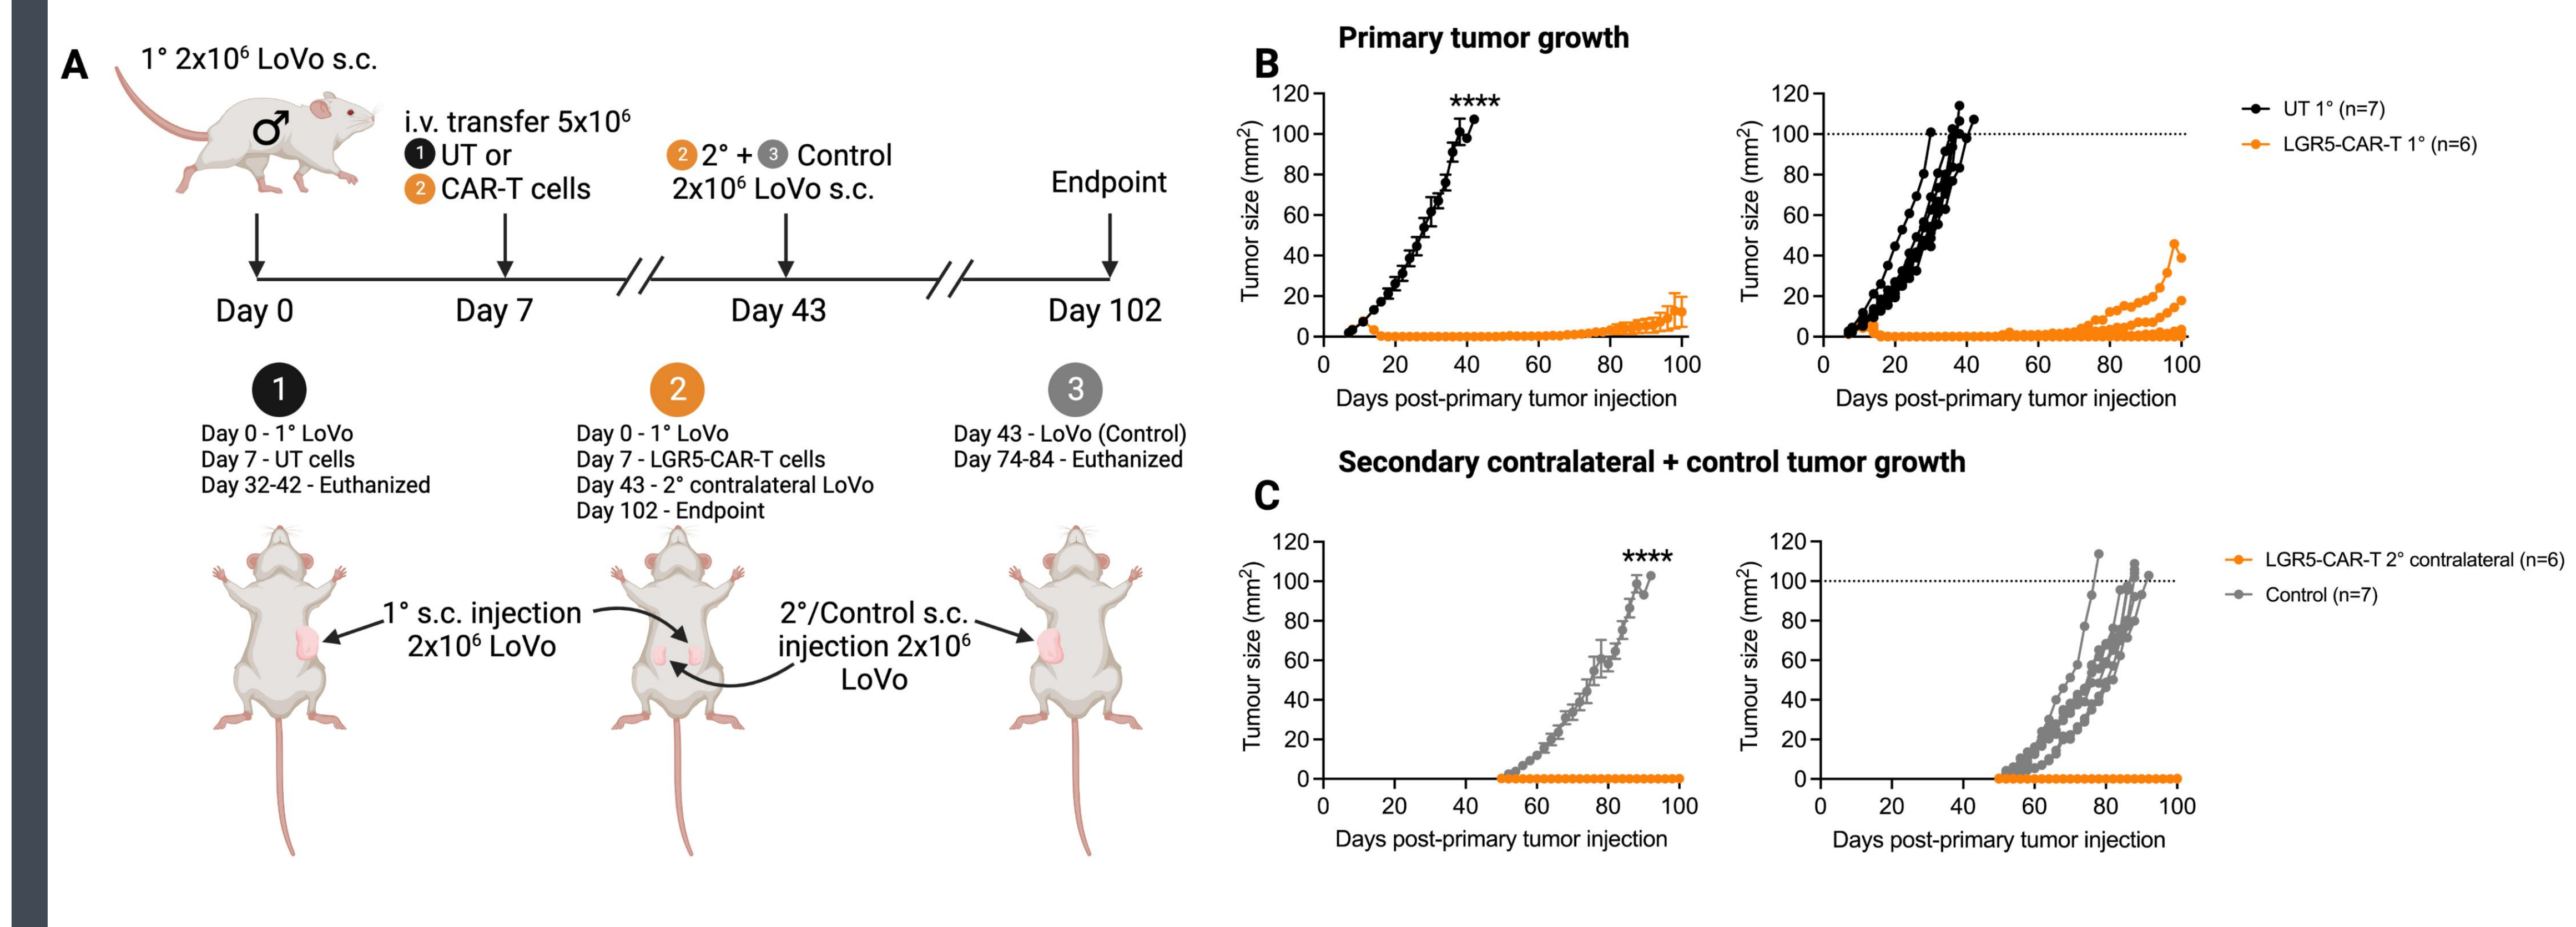

A
1° 2x10⁶ LoVo s.c.
i.v. transfer 5x10⁶ 1 UT or 2 CAR-T cells
2 2° + 3 Control 2x10⁶ LoVo s.c.
Endpoint
Day 0
Day 7
Day 43
Day 102
1 Day 0 - 1° LoVo Day 7 - UT cells Day 32-42 - Euthanized
2 Day 0 - 1° LoVo Day 7 - LGR5-CAR-T cells Day 43 - 2° contralateral LoVo Day 102 - Endpoint
3 Day 43 - LoVo (Control) Day 74-84 - Euthanized
1° s.c. injection 2x10⁶ LoVo
2°/Control s.c. injection 2x10⁶ LoVo
B
Primary tumor growth
UT 1° (n=7)
LGR5-CAR-T 1° (n=6)
C
Secondary contralateral + control tumor growth
LGR5-CAR-T 2° contralateral (n=6)
Control (n=7)
Tumor size (mm²)
Tumour size (mm²)
Days post-primary tumor injection

Figure 3. LGR5-targeting CAR-T cells reject primary tumors and provide lasting protection against tumor rechallenge. $5x10^6$ UT control or LGR5-targeting CAR-T cells were transferred i.v. day 7 post-primary tumor injection in LoVo-bearing male NSG mice. On day 43 post-primary tumour injection, a secondary $2x10^6$ LoVo s.c. challenge was administered to the contralateral flank of mice that received LGR5-targeting CAR-T cells. As a control, age-matched male NSG mice that had received no prior treatment were also injected with $2x10^6$ LoVo s.c.. Both primary and secondary tumor growth were assessed**.** **(A)** Experiment schematic. **(B)** Primary grouped (left) and individual (right) LoVo tumor growth curves. **(C)** Secondary contralateral and control grouped (left) and individual (right) LoVo tumor growth curves. Dotted line represents the $100mm^2$ tumor area euthanasia threshold. Data represented as the mean ± SEM; two-way ANOVA with Bonferroni's post-test ; ***p≤0.001, ****p≤0.0001.

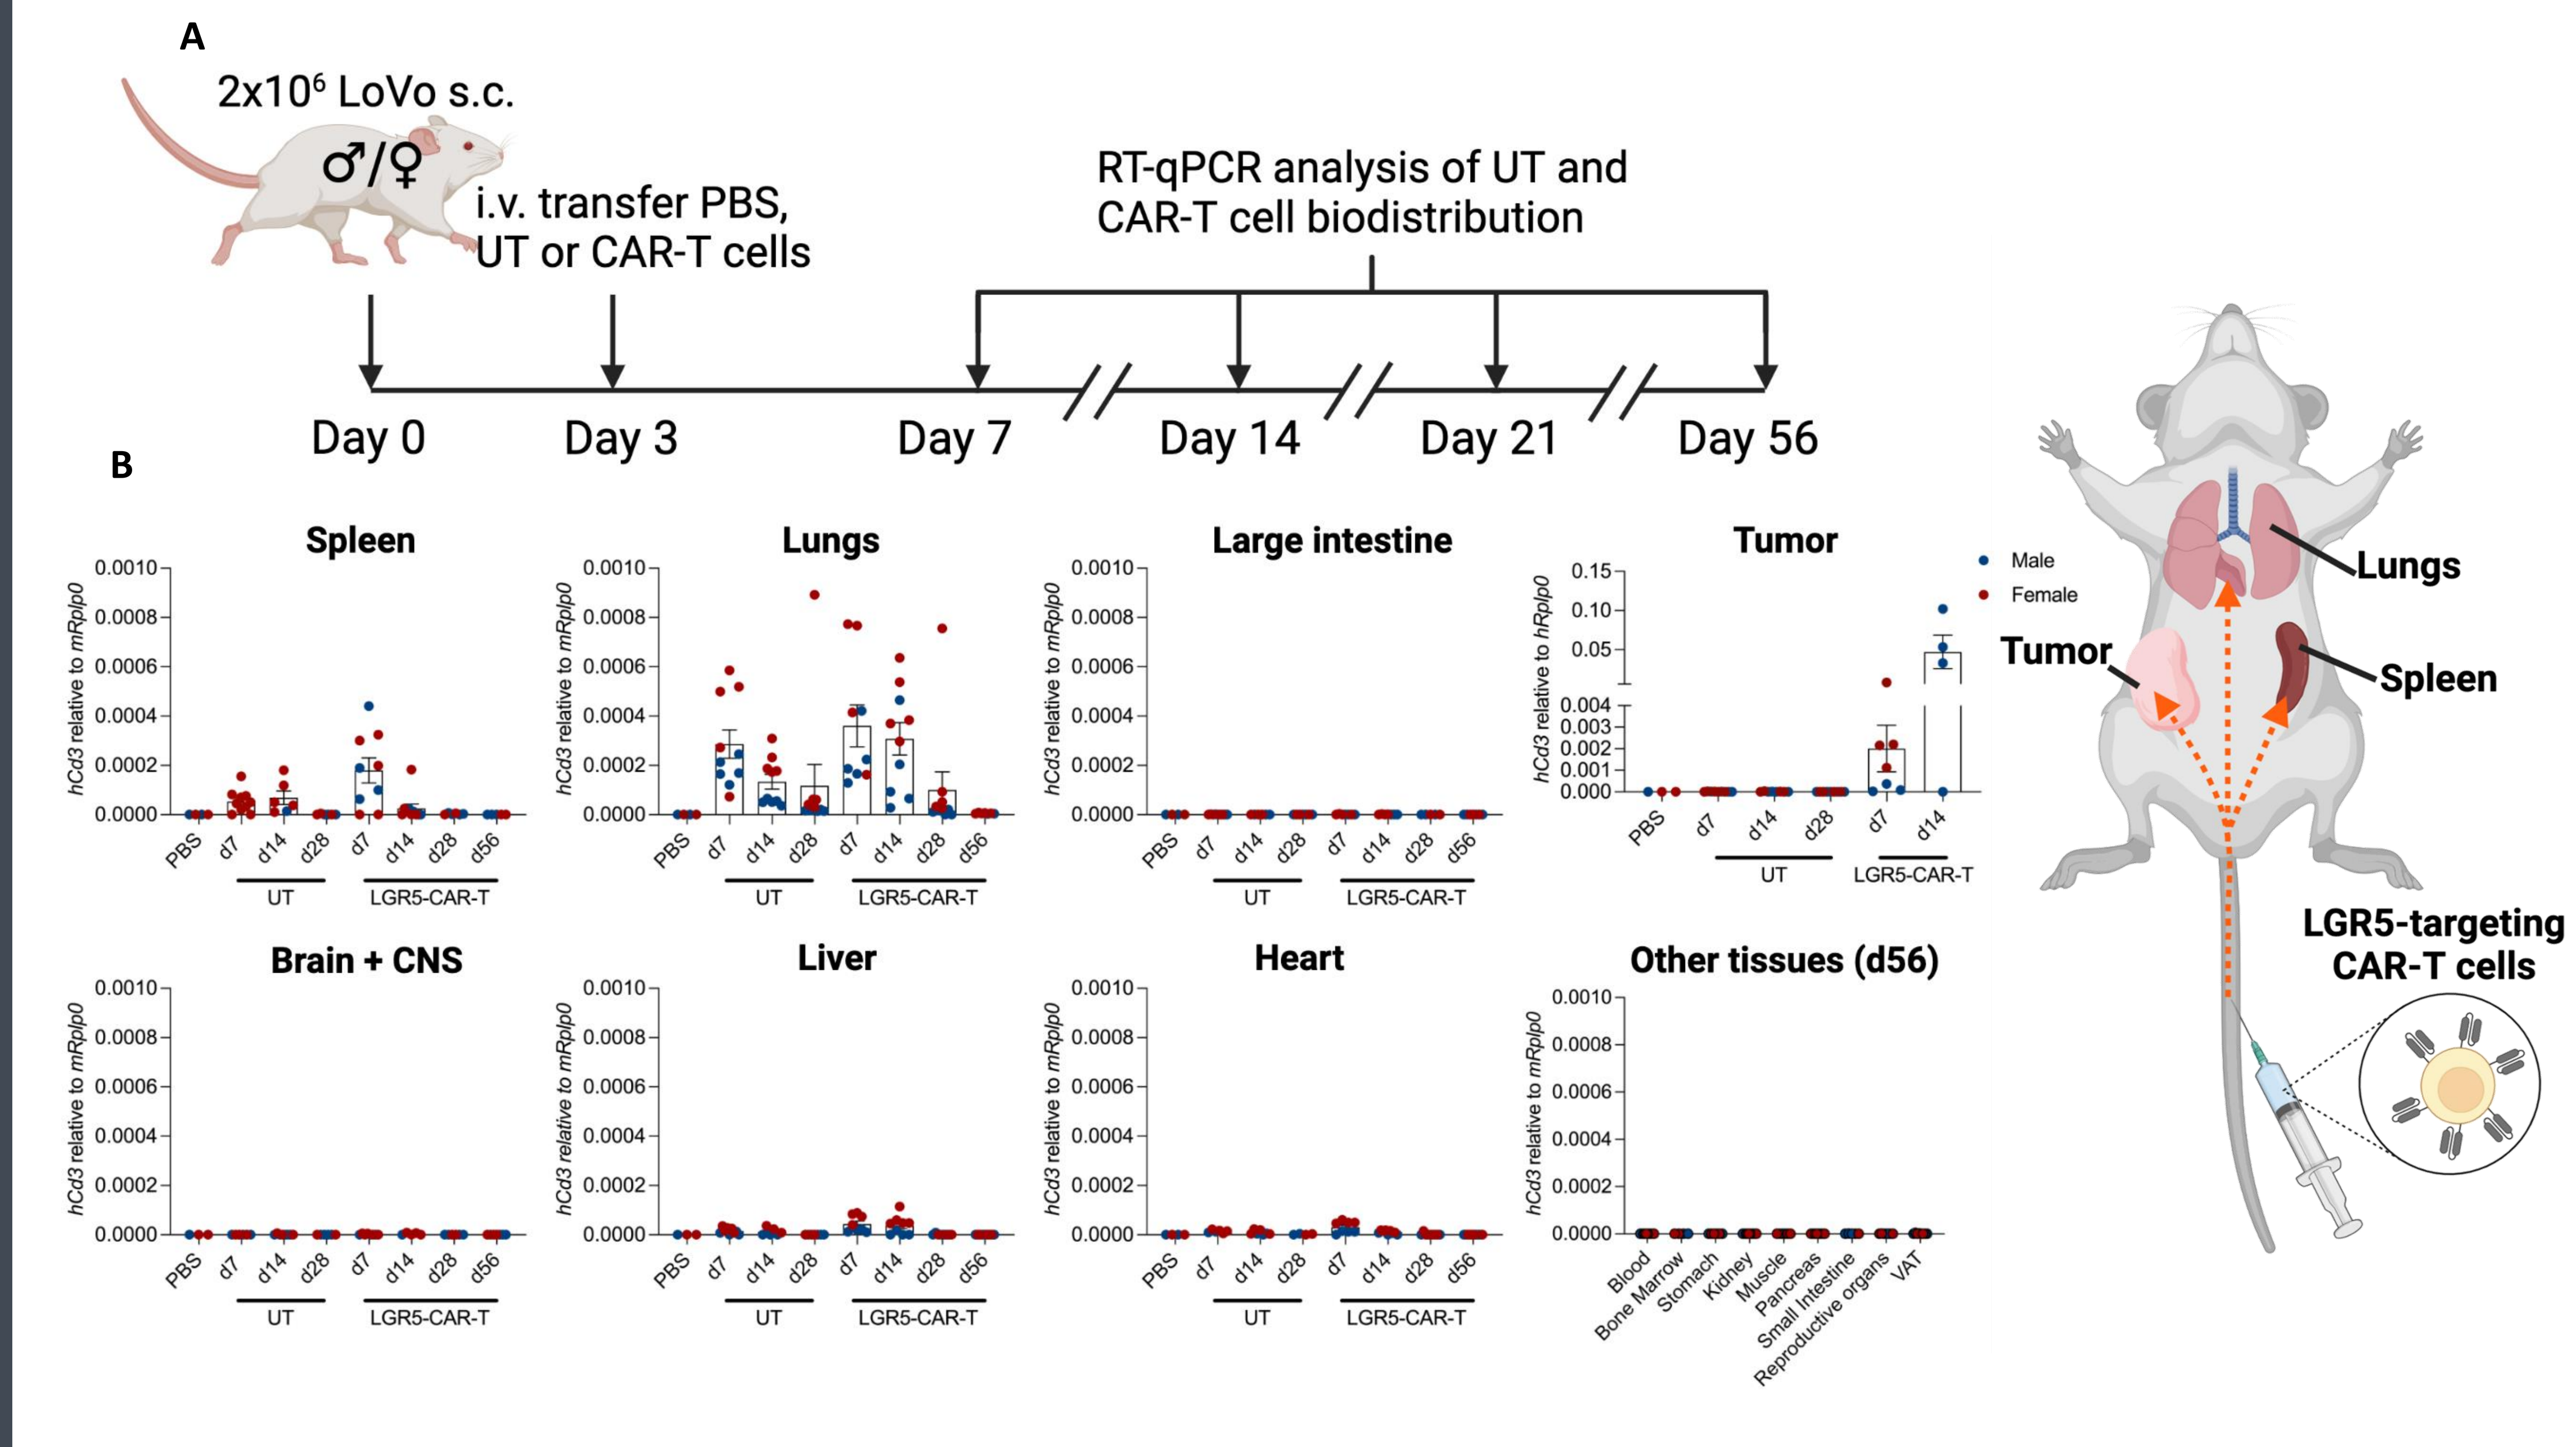

A
2x10⁶ LoVo s.c.
♂/♀
i.v. transfer PBS, UT or CAR-T cells
RT-qPCR analysis of UT and CAR-T cell biodistribution
Day 0
Day 3
Day 7
Day 14
Day 21
Day 56
B
Spleen
Lungs
Large intestine
Tumor
Brain + CNS
Liver
Heart
Other tissues (d56)
hCd3 relative to mRplp0
hCd3 relative to hRplp0
PBS
UT
LGR5-CAR-T
Male
Female
Blood
Bone Marrow
Stomach
Kidney
Muscle
Pancreas
Small Intestine
Reproductive organs
VAT
Lungs
Tumor
Spleen
LGR5-targeting CAR-T cells

Figure 4. *In vivo* spatiotemporal biodistribution analysis reveals that LGR5-targeting CAR-T cells disseminate to the tumor, lungs, and spleen. Male (blue symbols) and female (red symbols) NSG mice were injected s.c. with LoVo cells three days prior to intravenous transfer of $5x10^6$ LGR5-targeting CAR-T, UT T cells, or PBS. From separate groups of mice, sixteen tissues were harvested on day 7, 14, 21, or 56 post-tumor injection. RT-qPCR analysis measured the relative abundance of LGR5-targeting CAR-T and UT T cells over the course of the experiment. **(A)** Experiment schematic. **(B)** Relative abundance in the spleen, lungs, large intestine, tumor, brain, CNS, liver, and heart on days 7, 14, 28 and 56, and in other tissues on day-56. Data represented as expression of human *Cd3 (hCd3)* relative to mouse *Rplp0 (mRplp0)* to detect UT and LGR5-targeting CAR T cells in mouse organs (except for tumours where *hCd3* expression is relative to *hRplp0*). All data represented as the mean ± SEM; n = 4-10/group/timepoint.